108P

Preclinical Evaluation of Potential Therapeutic Combinations with the Kirsten Rat Sarcoma (KRAS)^{G12D} Inhibitor INCB161734 with Chemotherapy, Growth Factor Disruption, or Immunotherapy in Pancreatic Cancer

Presented at the

ESMO Targeted Anticancer Therapies Congress 2026

Paris, France • 16-18 March 2026

Matthew R. Farren,[1] **Valerie Roman**,[1] **Renee Wallower**,[1] **Alexandra Gallion**,[1] **Amanda Smith**,[1] **Hui Wang**,[1] **Sunkyu Kim**[1]

[1]Incyte Corporation, Wilmington, DE, USA

Abstract

Background: Kirsten Rat Sarcoma G12D (*KRAS*^{G12D}) mutation is found in ~40% of pancreatic ductal adenocarcinomas (PDACs); however, there are no approved agents targeting KRAS^{G12D}. INCB161734 is a potent, selective, and orally bioavailable KRAS^{G12D} inhibitor being assessed in a phase 1 study (NCT06179160). Early efficacy outcomes for KRAS^{G12D} inhibitors, including INCB161734, are encouraging, although fewer than half of patients with PDAC experience clinical response. We investigated whether INCB161734 combination therapies might improve therapeutic outcomes.

Methods: INCB161734-based combinations were tested in mice bearing subcutaneous PDAC tumours (xenograft and/or syngeneic). Combinations included INCB161734 with chemotherapy (gemcitabine + nab-paclitaxel [GEMNabP] or 5-fluorouracil + irinotecan + oxaliplatin + leucovorin [FOLFIRINOX]), the epidermal growth factor receptor (EGFR) antagonist cetuximab, or anti–programmed cell death protein 1 (PD-1)/programmed death-ligand 1 (PD-L1) monoclonal antibodies. Animal studies were conducted under the supervision of the INCYTE Institutional Animal Care and Use Committee.

Results: INCB161734 with either chemotherapy regimen improved antitumour activity compared with monotherapy. Chemotherapy combinations were more effective in immune-competent than immune-deficient mouse models, suggesting an immunomodulatory aspect of KRAS^{G12D} inhibition. INCB161734 + cetuximab resulted in greater tumour growth inhibition than monotherapy, likely by preventing EGFR-induced (re)activation of wild-type RAS signalling. Finally, combining INCB161734 with anti–PD-(L)1 antibodies resulted in complete or near-complete tumour regressions in 90% of treated animals, with 80% of animals remaining tumour-free for ≥60 days after the end of treatment, further suggesting an immunomodulatory impact of KRAS^{G12D} inhibition. All combinations were well tolerated.

Conclusions: These results demonstrate that KRAS^{G12D} blockade via INCB161734 combined with chemotherapy, EGFR signalling disruption, or immune checkpoint blockade via PD-1/PD-L1 may have the potential to improve therapeutic efficacy in patients with PDAC.

Introduction

- *KRAS* mutations are found in ~23% of all solid tumours, and *KRAS*^{G12D} mutations are the most common, occurring in ~40% of patients with PDAC and ~30% of patients with colorectal cancer[1]
- INCB161734 is a potent, selective, orally bioavailable KRAS^{G12D} inhibitor[2,3]
 - Picomolar binding affinity to KRAS^{G12D}, ~80-fold selectivity over wild-type KRAS
 - Equal binding affinity to both GDP (RAS-OFF) and GTP (RAS-ON) forms of KRAS^{G12D}
 - Inhibits KRAS^{G12D} pathway and proliferation of KRAS^{G12D} mutant cells with nanomolar potency in *in vitro* cellular assays
 - Highly orally bioavailable in preclinical studies, supporting exposure >IC_{90} at trough concentration (C_{trough})[4]
 - Well tolerated in preclinical studies
 - In clinical development (NCT06179160)

INCB161734 Antitumour Activity Is Enhanced When Combined with PDAC Chemotherapy

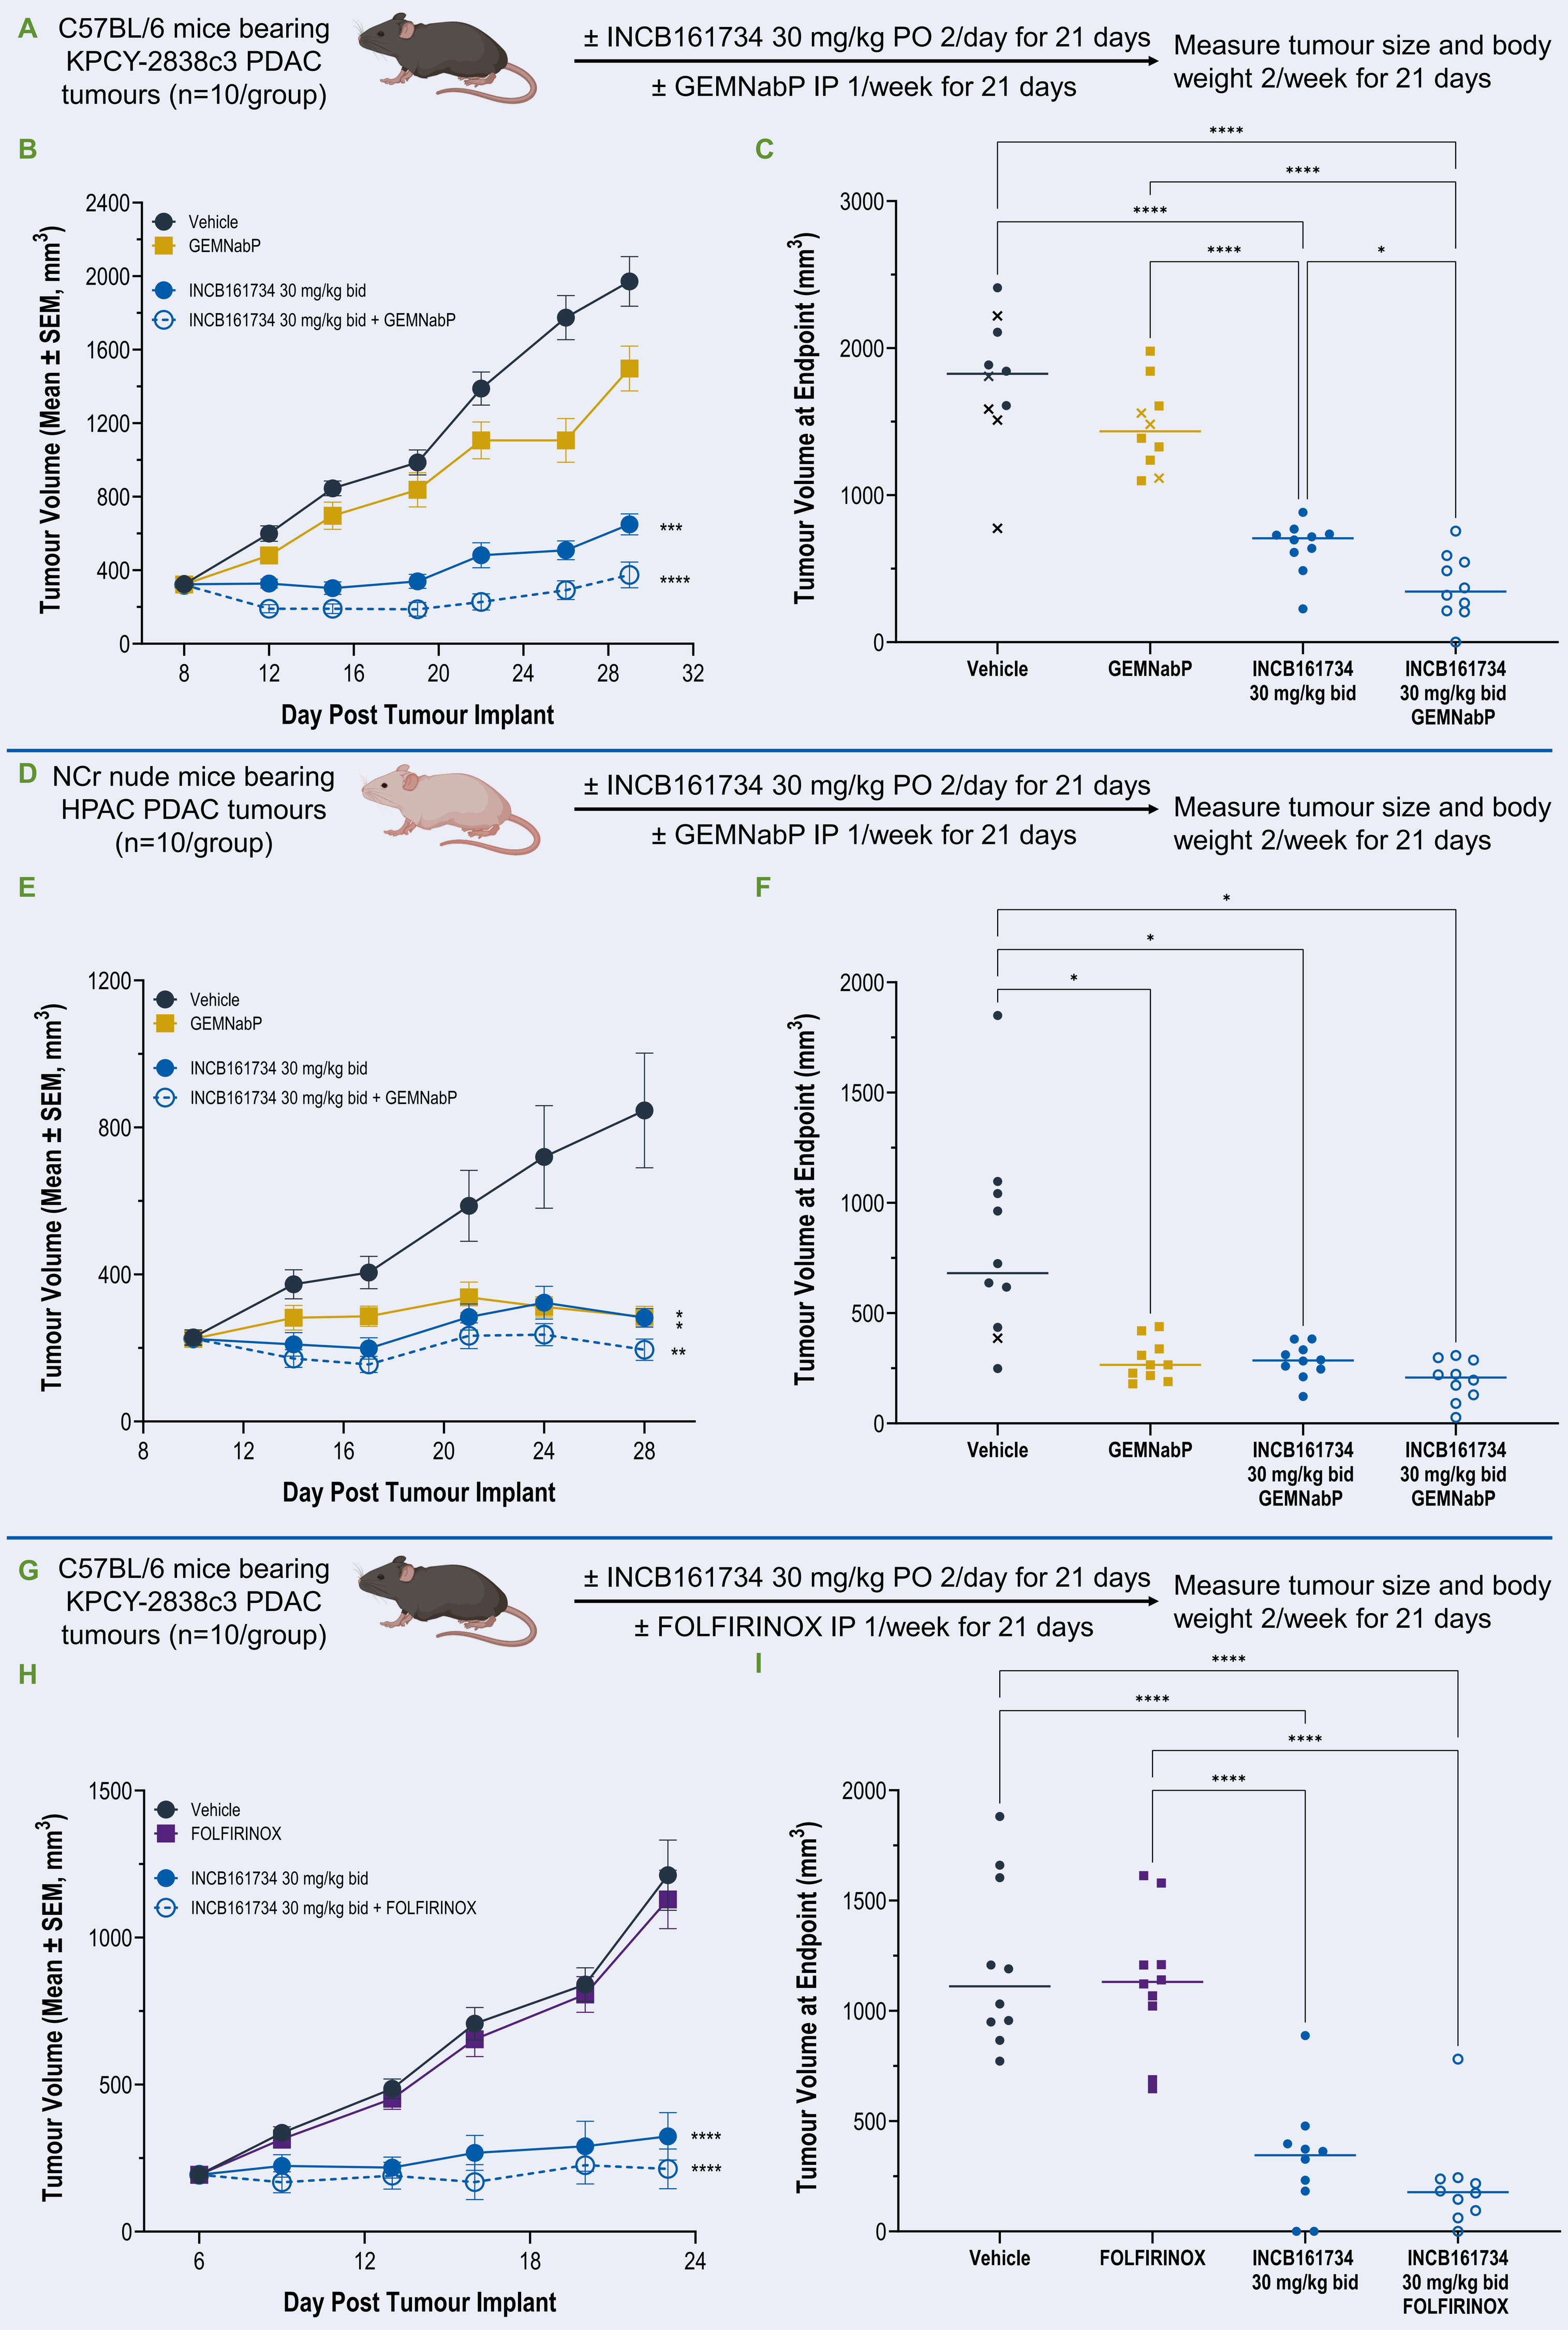
A C57BL/6 mice bearing KPCY-2838c3 PDAC tumours (n=10/group)
± INCB161734 30 mg/kg PO 2/day for 21 days
± GEMNabP IP 1/week for 21 days
Measure tumour size and body weight 2/week for 21 days
B
Vehicle
GEMNabP
INCB161734 30 mg/kg bid
INCB161734 30 mg/kg bid + GEMNabP
Tumour Volume (Mean ± SEM, mm³)
Day Post Tumour Implant
C
Tumour Volume at Endpoint (mm³)
Vehicle
GEMNabP
INCB161734 30 mg/kg bid
INCB161734 30 mg/kg bid GEMNabP
D NCr nude mice bearing HPAC PDAC tumours (n=10/group)
± INCB161734 30 mg/kg PO 2/day for 21 days
± GEMNabP IP 1/week for 21 days
Measure tumour size and body weight 2/week for 21 days
E
F
G C57BL/6 mice bearing KPCY-2838c3 PDAC tumours (n=10/group)
± INCB161734 30 mg/kg PO 2/day for 21 days
± FOLFIRINOX IP 1/week for 21 days
Measure tumour size and body weight 2/week for 21 days
H
Vehicle
FOLFIRINOX
INCB161734 30 mg/kg bid
INCB161734 30 mg/kg bid + FOLFIRINOX
I
INCB161734 30 mg/kg bid FOLFIRINOX

GEMNabP – Gemcitabine (30 mg/kg) + Nab-paclitaxel (100 mg/kg) formulated in sterile saline and administered once weekly via intraperitoneal (IP) injection; FOLFORINOX – 5-fluorouracil (25 mg/kg) + oxaliplatin (2.5 mg/kg) + irinotecan (25 mg/kg) + folinic acid (50 mg/kg) formulated in sterile saline and administered once weekly via IP injection. * *P*<0.05; ** *P*<0.01; *** *P*<0.0005; **** *P*<0.0001. Statistical comparisons via two-way ANOVA in B, E, and H are between the indicated group and the vehicle control group. Statistical comparisons via one-way ANOVA in C, F, and I are between the indicated groups. bid, twice daily; HPAC, human pancreatic adenocarcinoma cells; PO, orally; SEM, standard error of the mean.

- The KRAS^{G12D} inhibitor INCB161734 exerted significant antitumour activity against *KRAS*^{G12D} mutant PDAC tumours as a monotherapy (A-I), and this was further enhanced when combined with PDAC standard-of-care chemotherapy regimens GEMNabP (A-F) or FOLFIRINOX (G-I)
 - (A-C) Syngeneic C57BL/6 mice bearing KPCY-2838c3 tumours were treated ± INCB161734 ± GEMNabP
 - (A) Study design, (B) tumour growth over time, and (C) tumour size at endpoint
 - Note that "X" symbols denote mice that were removed from study early due to tumour ulceration
 - (D-F) NCr nude mice bearing HPAFII xenograft tumours were treated ± INCB161734 ± GEMNabP
 - (D) Study design, (E) tumour growth over time, and (F) tumour size at endpoint
 - Note that "X" symbols denote mice that were removed from study early due to tumour ulceration
 - (G-I) Syngeneic C57BL/6 mice bearing KPCY-2838c3 tumours were treated ± INCB161734 ± FOLFIRINOX
 - (G) Study design, (H) tumour growth over time, and (I) tumour size at endpoint
- Both combinations (INCB161734 + GEMNabP and INCB161734 + FOLFIRINOX) were well tolerated, without significant weight loss or other clinical tolerability signals

Combination INCB161734 + Cetuximab (EGFRi) Generates Superior Tumour Growth Inhibition

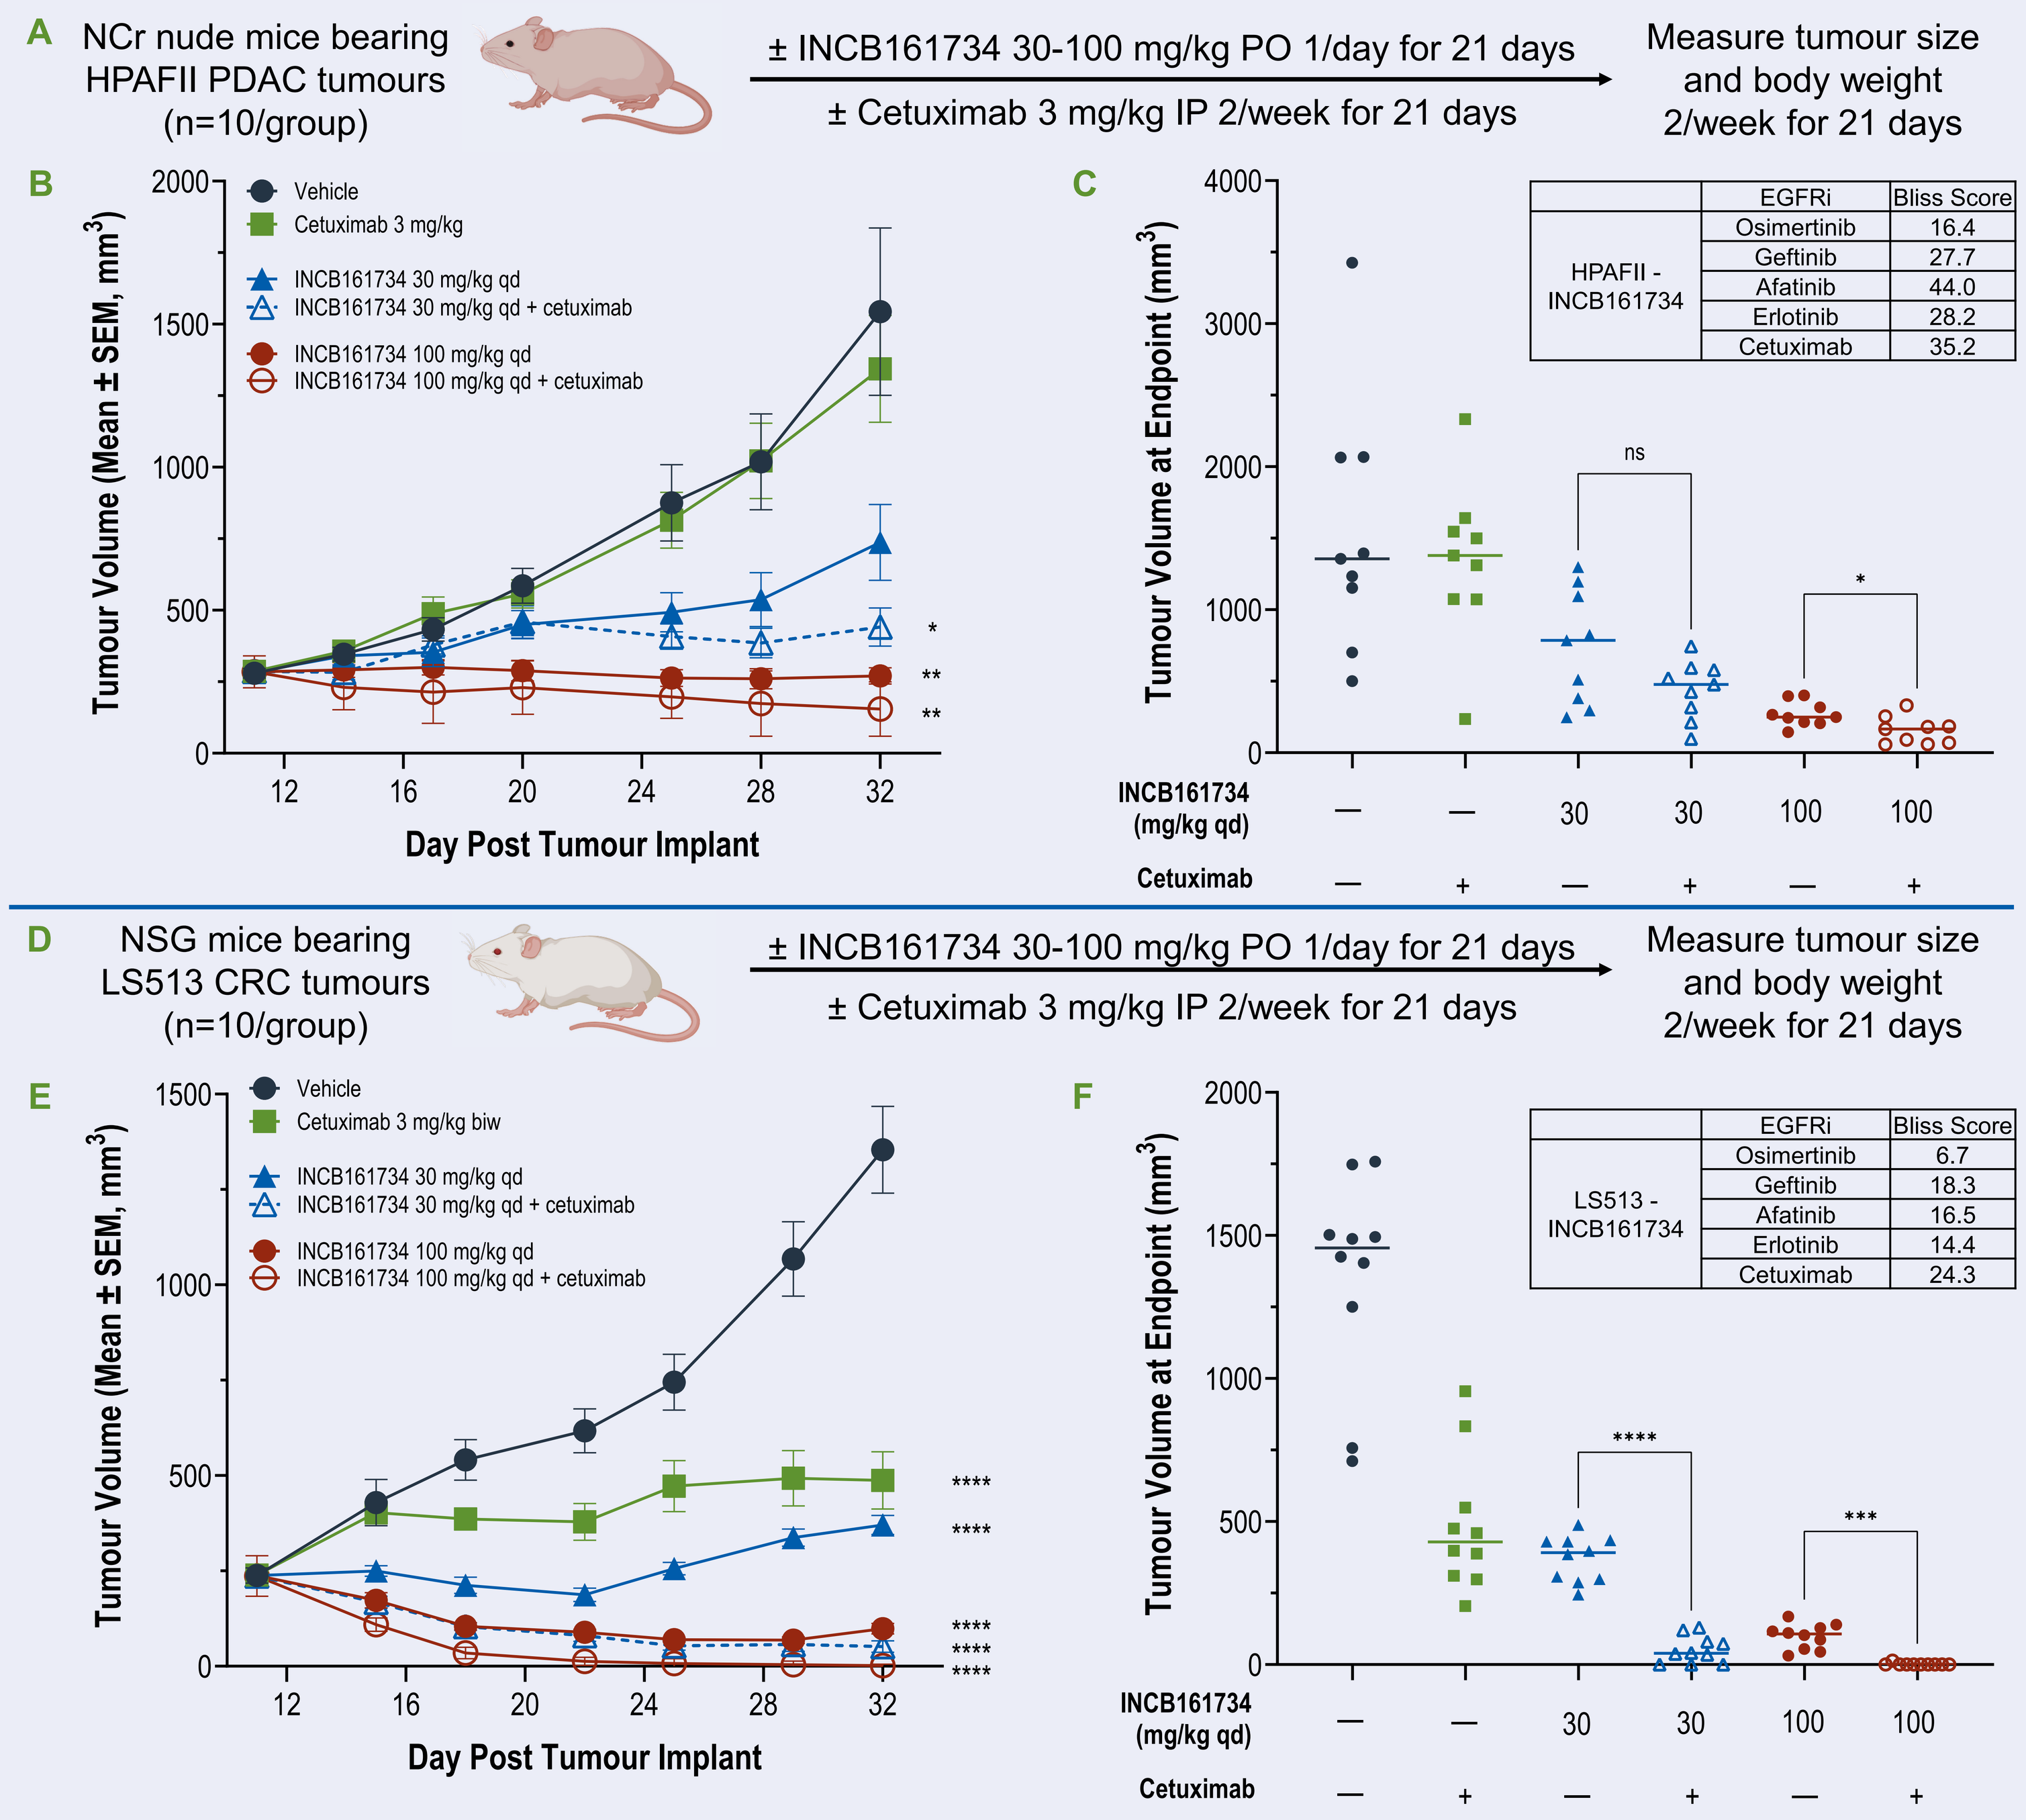
A NCr nude mice bearing HPAFII PDAC tumours (n=10/group)
± INCB161734 30-100 mg/kg PO 1/day for 21 days
± Cetuximab 3 mg/kg IP 2/week for 21 days
Measure tumour size and body weight 2/week for 21 days
B
Vehicle
Cetuximab 3 mg/kg
INCB161734 30 mg/kg qd
INCB161734 30 mg/kg qd + cetuximab
INCB161734 100 mg/kg qd
INCB161734 100 mg/kg qd + cetuximab
Tumour Volume (Mean ± SEM, mm³)
Day Post Tumour Implant
C
Tumour Volume at Endpoint (mm³)
HPAFII - INCB161734
EGFRi | Bliss Score
Osimertinib 16.4
Geftinib 27.7
Afatinib 44.0
Erlotinib 28.2
Cetuximab 35.2
INCB161734 (mg/kg qd)
Cetuximab
D NSG mice bearing LS513 CRC tumours (n=10/group)
± INCB161734 30-100 mg/kg PO 1/day for 21 days
± Cetuximab 3 mg/kg IP 2/week for 21 days
Measure tumour size and body weight 2/week for 21 days
E
Cetuximab 3 mg/kg biw
F
LS513 - INCB161734
Osimertinib 6.7
Geftinib 18.3
Afatinib 16.5
Erlotinib 14.4
Cetuximab 24.3

Cetuximab was diluted in saline and administered twice weekly via IP injection. * *P*<0.05; ** *P*<0.01; *** *P*<0.0005; **** *P*<0.0001. Statistical comparisons via two-way ANOVA in B and E are between the indicated group and the vehicle control group. Statistical comparisons via one-way ANOVA in C and F are between the indicated groups. BLISS scores were calculated using SynergyFinder and values represent the "most synergistic area." ANOVA, analysis of variance; biw, twice weekly; CRC, colorectal cancer; EGFRi, epidermal growth factor receptor inhibitor; ns, not significant; qd, once daily; SEM, standard error of the mean.

- The antitumour activity of INCB161734 in PDAC (A-C) and CRC (D-F) was significantly increased by combination with the EGFR inhibitor cetuximab
 - (A-C) NCr nude mice bearing HPAFII xenograft tumours were treated ± INCB161734 ± cetuximab
 - (A) Study design, (B) tumour growth over time, and (C) tumour size at endpoint, including *in vitro* Synergy Score testing EGFR inhibitors in combination with INCB161734 in HPAFII cells
 - (D-F) NSG mice bearing LS513 xenograft tumours were treated ± INCB161734 ± GEMNabP
 - (D) Study design, (E) tumour growth over time, and (F) tumour size at endpoint, including *in vitro* Synergy Score testing EGFR inhibitors in combination with INCB161734 in LS513 cells
- This combination was well tolerated, without significant weight loss or other clinical tolerability signals

INCB161734 Potentiates Immune Checkpoint Blockade, Generating Durable Antitumour Activity

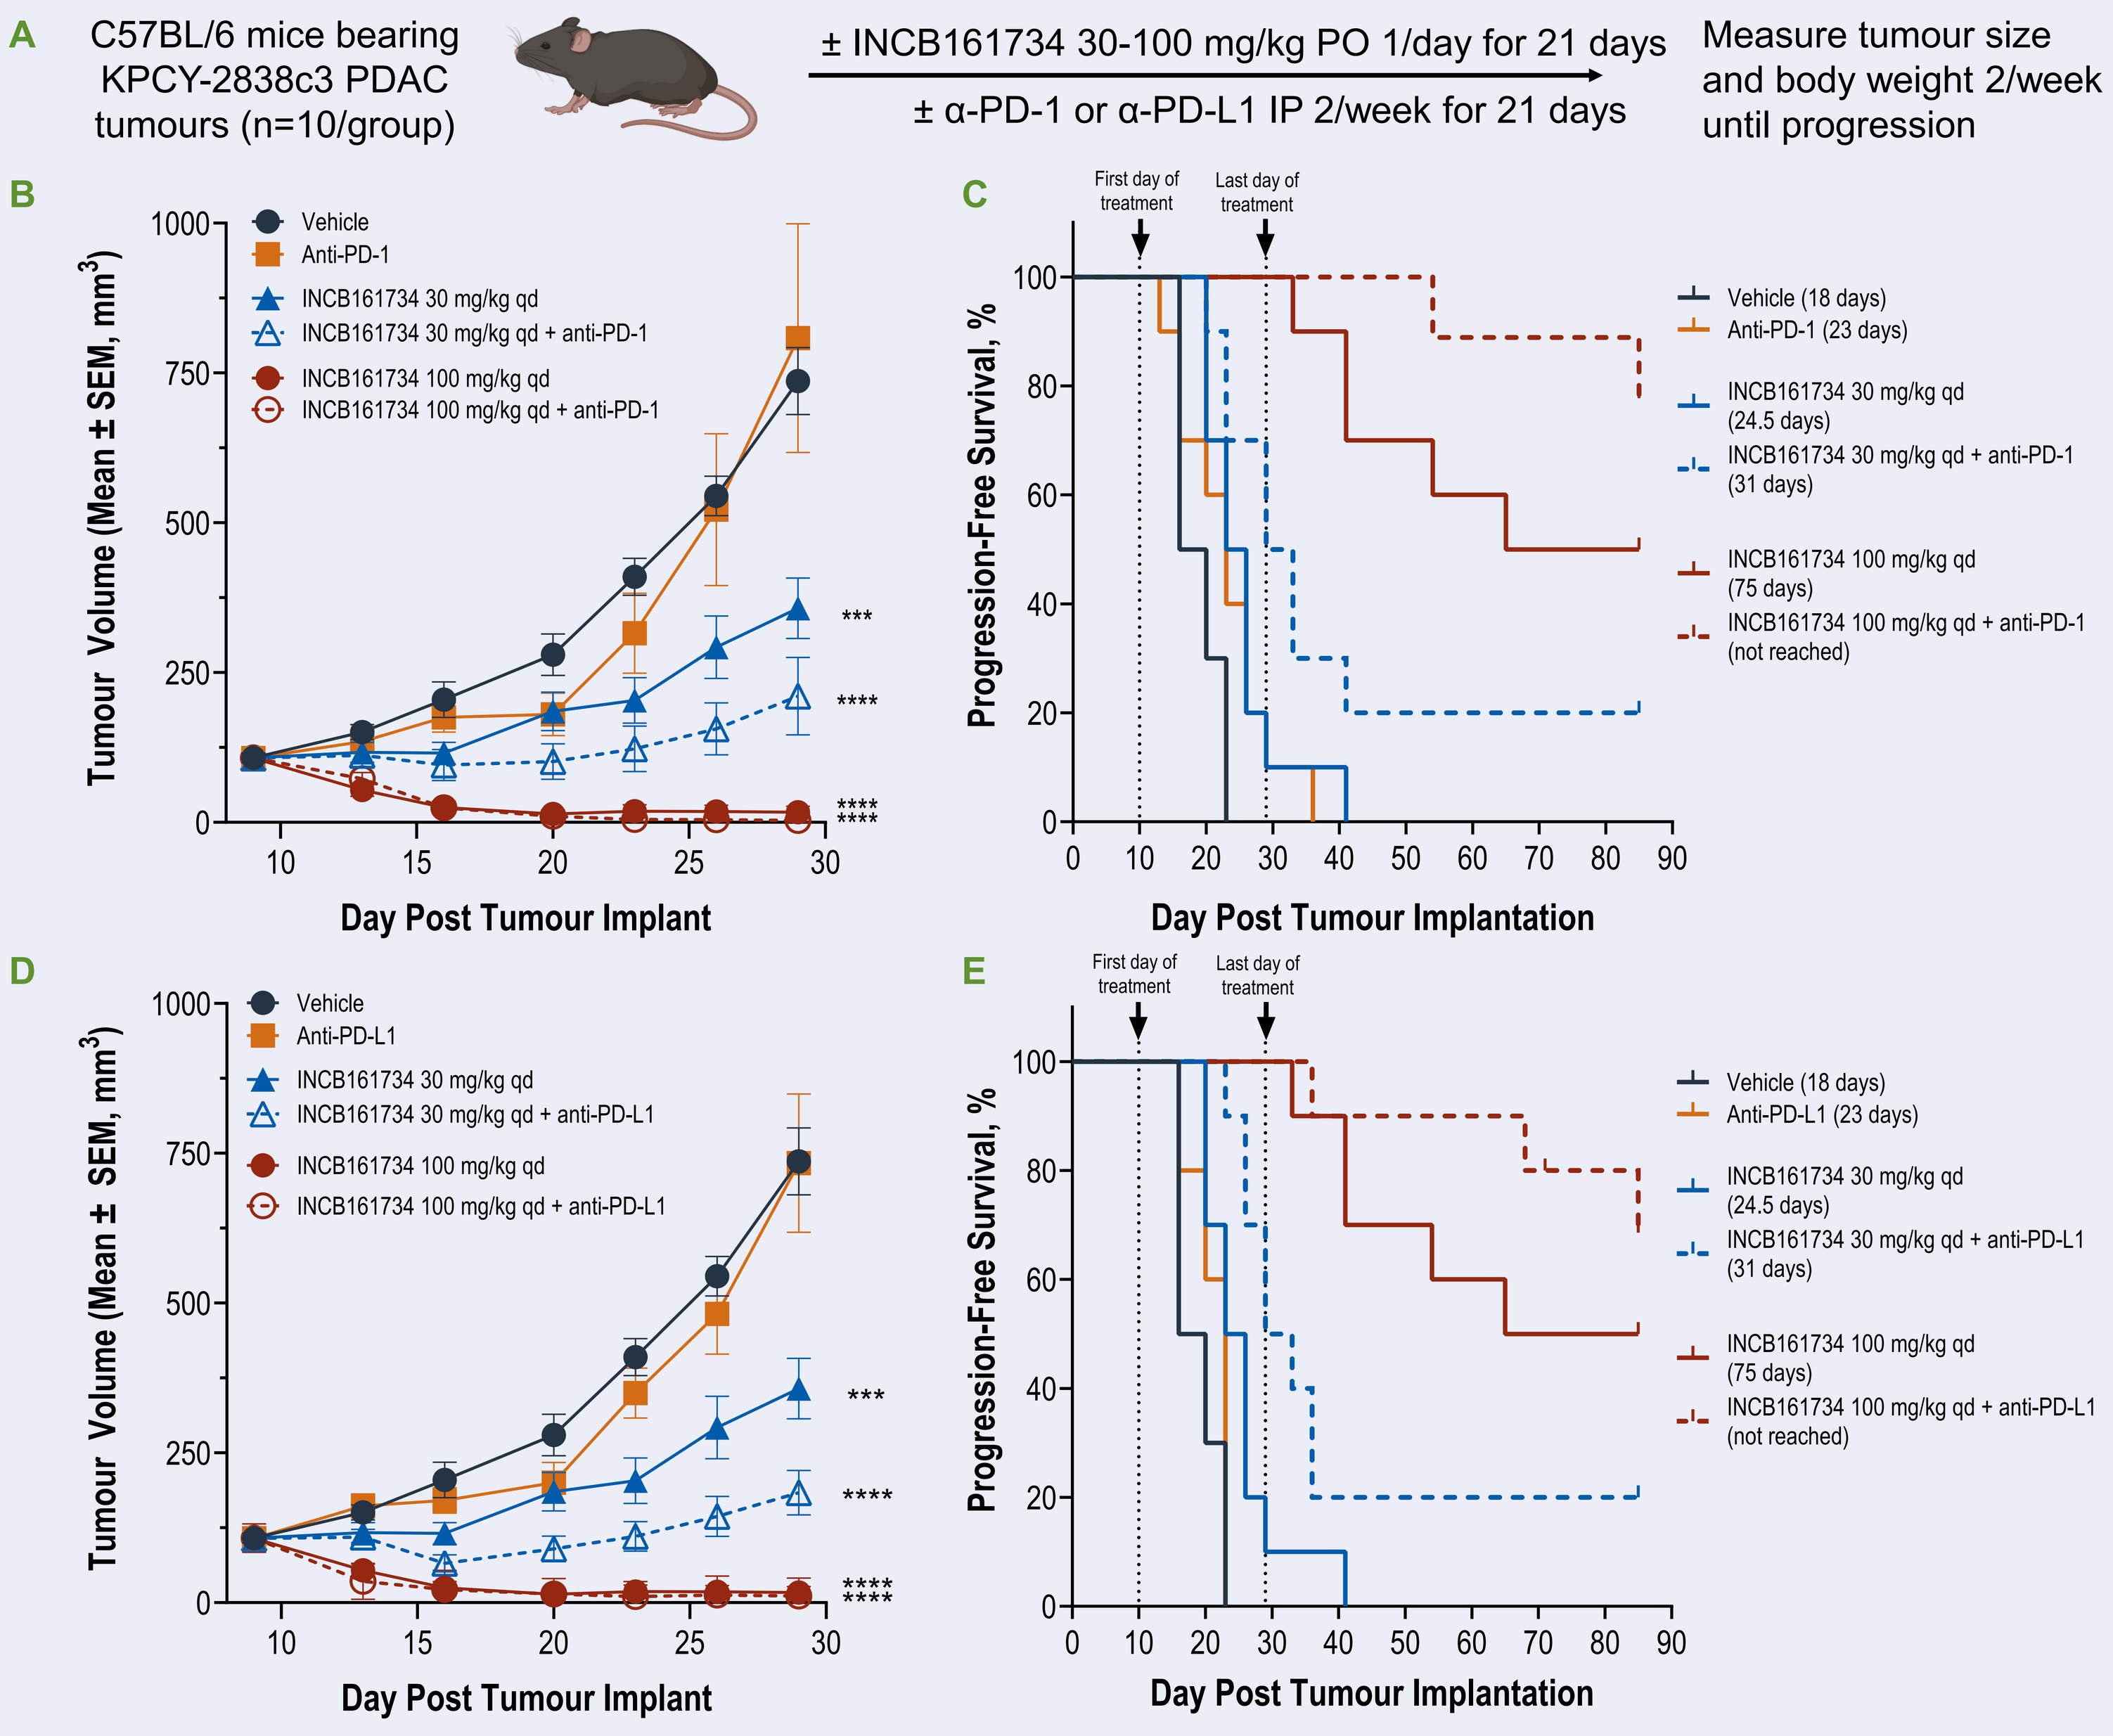
A C57BL/6 mice bearing KPCY-2838c3 PDAC tumours (n=10/group)
± INCB161734 30-100 mg/kg PO 1/day for 21 days
± α-PD-1 or α-PD-L1 IP 2/week for 21 days
Measure tumour size and body weight 2/week until progression
B
Vehicle
Anti-PD-1
INCB161734 30 mg/kg qd
INCB161734 30 mg/kg qd + anti-PD-1
INCB161734 100 mg/kg qd
INCB161734 100 mg/kg qd + anti-PD-1
Tumour Volume (Mean ± SEM, mm³)
Day Post Tumour Implant
C
First day of treatment
Last day of treatment
Progression-Free Survival, %
Vehicle (18 days)
Anti-PD-1 (23 days)
INCB161734 30 mg/kg qd (24.5 days)
INCB161734 30 mg/kg qd + anti-PD-1 (31 days)
INCB161734 100 mg/kg qd (75 days)
INCB161734 100 mg/kg qd + anti-PD-1 (not reached)
Day Post Tumour Implantation
D
Anti-PD-L1
INCB161734 30 mg/kg qd + anti-PD-L1
INCB161734 100 mg/kg qd + anti-PD-L1
E
Anti-PD-L1 (23 days)
INCB161734 30 mg/kg qd + anti-PD-L1 (31 days)
INCB161734 100 mg/kg qd + anti-PD-L1 (not reached)

α-PD-1 (Clone BE0146) was diluted in saline and administered twice weekly via IP injection; α-PD-L1 (Clone BE0101) was diluted in saline and administered twice weekly via IP injection. Tumour progression defined as tumour volume doubling compared with initial day of treatment. *** *P*<0.0005; **** *P*<0.0001. Statistical comparisons via two-way ANOVA in B are between the indicated group and the vehicle control group. α-PD-1, anti-programmed death-1 antibody; α-PD-L1, anti-programmed death-ligand 1; ANOVA, analysis of variance; IP, intraperitoneal; qd, once daily.

- (A) Study design. C57BL/6 mice bearing syngeneic KPCY-2838c3 tumours were treated ± INCB161734 ± α-PD-1 (B-C) or α-PD-L1 (D-E)
- Combining INCB161734 and immune checkpoint blockade generates deeper and more durable immune responses, with more complete tumour regression or slowing during the dosing period and more animals remaining tumour-free for months after the end of treatment
- These combinations were well tolerated, without significant weight loss or other clinical tolerability signals

Conclusions

- **INCB161734 is a novel, potent, selective, and orally bioavailable KRAS^{G12D} small molecule inhibitor that demonstrates considerable monotherapy activity in preclinical models**
- **This monotherapy activity can be further enhanced when INCB161734 is combined with other treatment modalities**
 - **Combinations with PDAC standard-of-care chemotherapy regimens generated significant additional efficacy over INCB161734 alone**
 - **Combinations with EGFR-blocking agents, such as cetuximab, drove significant antitumour activity, including partial or complete tumour regressions**
 - **KRAS^{G12D} blockade with INCB161734 in combination with immune checkpoint (anti–PD-1/PD-L1) blockade generated durable responses, with most animals (at the high dose) remaining tumour-free for more than 60 days after treatment ended**
- **INCB161734 was well tolerated as a monotherapy in preclinical models, and all the combinations described here were also well tolerated, with no evidence of meaningful weight loss or other tolerability signals**
- **These results demonstrate that combination therapies have the potential to improve therapeutic efficacy in PDAC**
- **INCB161734 is currently undergoing clinical evaluation as both monotherapy and in combination therapy settings (NCT06179160)**

Disclosures

Farren, Roman, Wallower, Gallion, Smith, Wang, Kim: Employment and stock ownership – Incyte Corporation.

Acknowledgements

All experiments carried out with funding by Incyte Corporation, Wilmington, DE, USA. Authors would like to thank all colleagues who contributed to this research effort. Editorial and graphics support was provided by Envision Pharma Group (Fairfield, CT, USA) and funded by Incyte Corporation.

References

1. Lee JK, et al. *NPJ Precis Oncol.* 2022;6:91. **2.** Farren MR, et al. AACR 2024. Abstract 5900. **3.** Wainberg Z, et al. ASCO GI 2026. Abstract 654. **4.** Incyte Corporation. Data on file.

To download a copy of this poster, scan code